Understanding Patient Experience in the Management and Prioritization of MASH and Comorbidities: Results from a Claims Analysis and Qualitative Patient Interviews

Jamie Hadac, PhD,[1] Ashwin Anand, MPH,[2] Julia Jones, BA,[3] Daniel Neilson, BS,[2] Wouter van der Pluijm, MPH,[2] Audrey Wu, MBA,[3] Leslie Fok, MPH[1]

[1]89bio, San Francisco, CA, USA; [2]Forian, Newtown, PA, USA; [3]ThinkGen, Wynnewood, PA, USA

SUMMARY

- Patients with metabolic dysfunction-associated steatohepatitis (MASH) often have several other health conditions.
- In a claims-based analysis, > 50% of patients with F2/F3 MASH had ≥ 3 comorbidities, the most common being high blood pressure, high cholesterol, diabetes, and obesity.
- Qualitative interviews with people living with MASH revealed that there are often delays in MASH diagnosis and management, as both physicians and patients may focus on other health issues first.
- Patients can feel confused or frustrated when physicians explain their MASH diagnosis too simplistically or dismissively.
- Managing MASH alongside multiple comorbidities is difficult; patients often face challenges coordinating their own care and accessing medications.

BACKGROUND

- MASH is a more advanced form of metabolic dysfunction-associated steatotic liver disease, which is increasing in prevalence globally.[1,2]
- Patients with MASH often have other comorbid metabolic conditions.[3]
- Due to a clinical focus on other comorbidities and the historic lack of effective treatments for MASH, MASH may be overlooked, resulting in delays in diagnosis, referral, and treatment.[4]
- To understand the burden of comorbidities and how people living with MASH prioritize its management relative to other diagnoses, we conducted a claims analysis and qualitative patient interviews.

METHODS

- Adults (> 18 years old in 2024) with MASH and F2 or F3 fibrosis were identified in Forian's CHRONOS™ data ecosystem from January 2023 to December 2024 based on International Classification of Diseases, Tenth Revision (ICD-10) and Logical Observation Identifiers Names and Codes (LOINC) codes.
 - The CHRONOS™ data ecosystem is compliant with the Health Insurance Portability and Accountability Act and comprises de-identified patient-level healthcare claims and electronic health record data.[5]
 - Patients with ICD-10 codes for alcoholic fatty liver disease/hepatitis/fibrosis/cirrhosis or liver cancers were excluded.
 - Descriptive statistics were used to analyze patient characteristics and comorbidity burden (based on custom grouping of ICD-10 codes with aggregated codes for obesity, hypertension, and hyperlipidemia; COVID-19 and 'Z' [encounter] codes were excluded).
- Qualitative, web-assisted interviews with 14 people living with MASH were conducted in two phases, from December 2024 to January 2025 (1-hour interviews) and from August to September 2025 (30-minute interviews).
 - All participants provided written informed consent.
 - Interview transcripts and recordings were reviewed for common themes.

RESULTS

Table 1. Demographics of patients with F2/F3 MASH identified in the claims analysis

Parameter	Patients with F2/F3 MASH (N = 5473)
Sex, n (%)	
Male	2310 (42.2)
Female	3036 (55.5)
Unknown	127 (2.3)
Age group, years, n (%)	
18–19	1 (< 0.1)
20–29	32 (0.6)
30–39	97 (1.8)
40–49	350 (6.4)
50–59	1220 (22.3)
60–69	1900 (34.7)
70–79	1280 (23.4)
≥ 80	473 (8.6)
Unknown	120 (2.2)

MASH, metabolic dysfunction-associated steatohepatitis.

CLAIMS ANALYSIS

- In total, 5473 patients with F2/F3 MASH were identified; 55% were female (**Table 1**).
- Of these patients, 87% had ≥ 1 comorbidity, 75% had ≥ 2 comorbidities, and 55% had ≥ 3 comorbidities (**Figure 1**).
- The most common comorbidities were hypertensive conditions (67%), dyslipidemia (63%), diabetes (48%), and obesity (43%) (**Figure 2**).
- Beyond cardiometabolic conditions, 29% of patients had osteoarthritis, 22% had anxiety, 15% had depression, and 12% had asthma.

INSIGHTS FROM QUALITATIVE PATIENT INTERVIEWS

- The mean age of the interviewed individuals with MASH was 52 years (range 28–74 years) and the majority had F2 or F3 fibrosis (**Table 2**).

RESULTS (CONTINUED)

Figure 1. Number of comorbidities among patients with F2/F3 MASH identified in the claims analysis

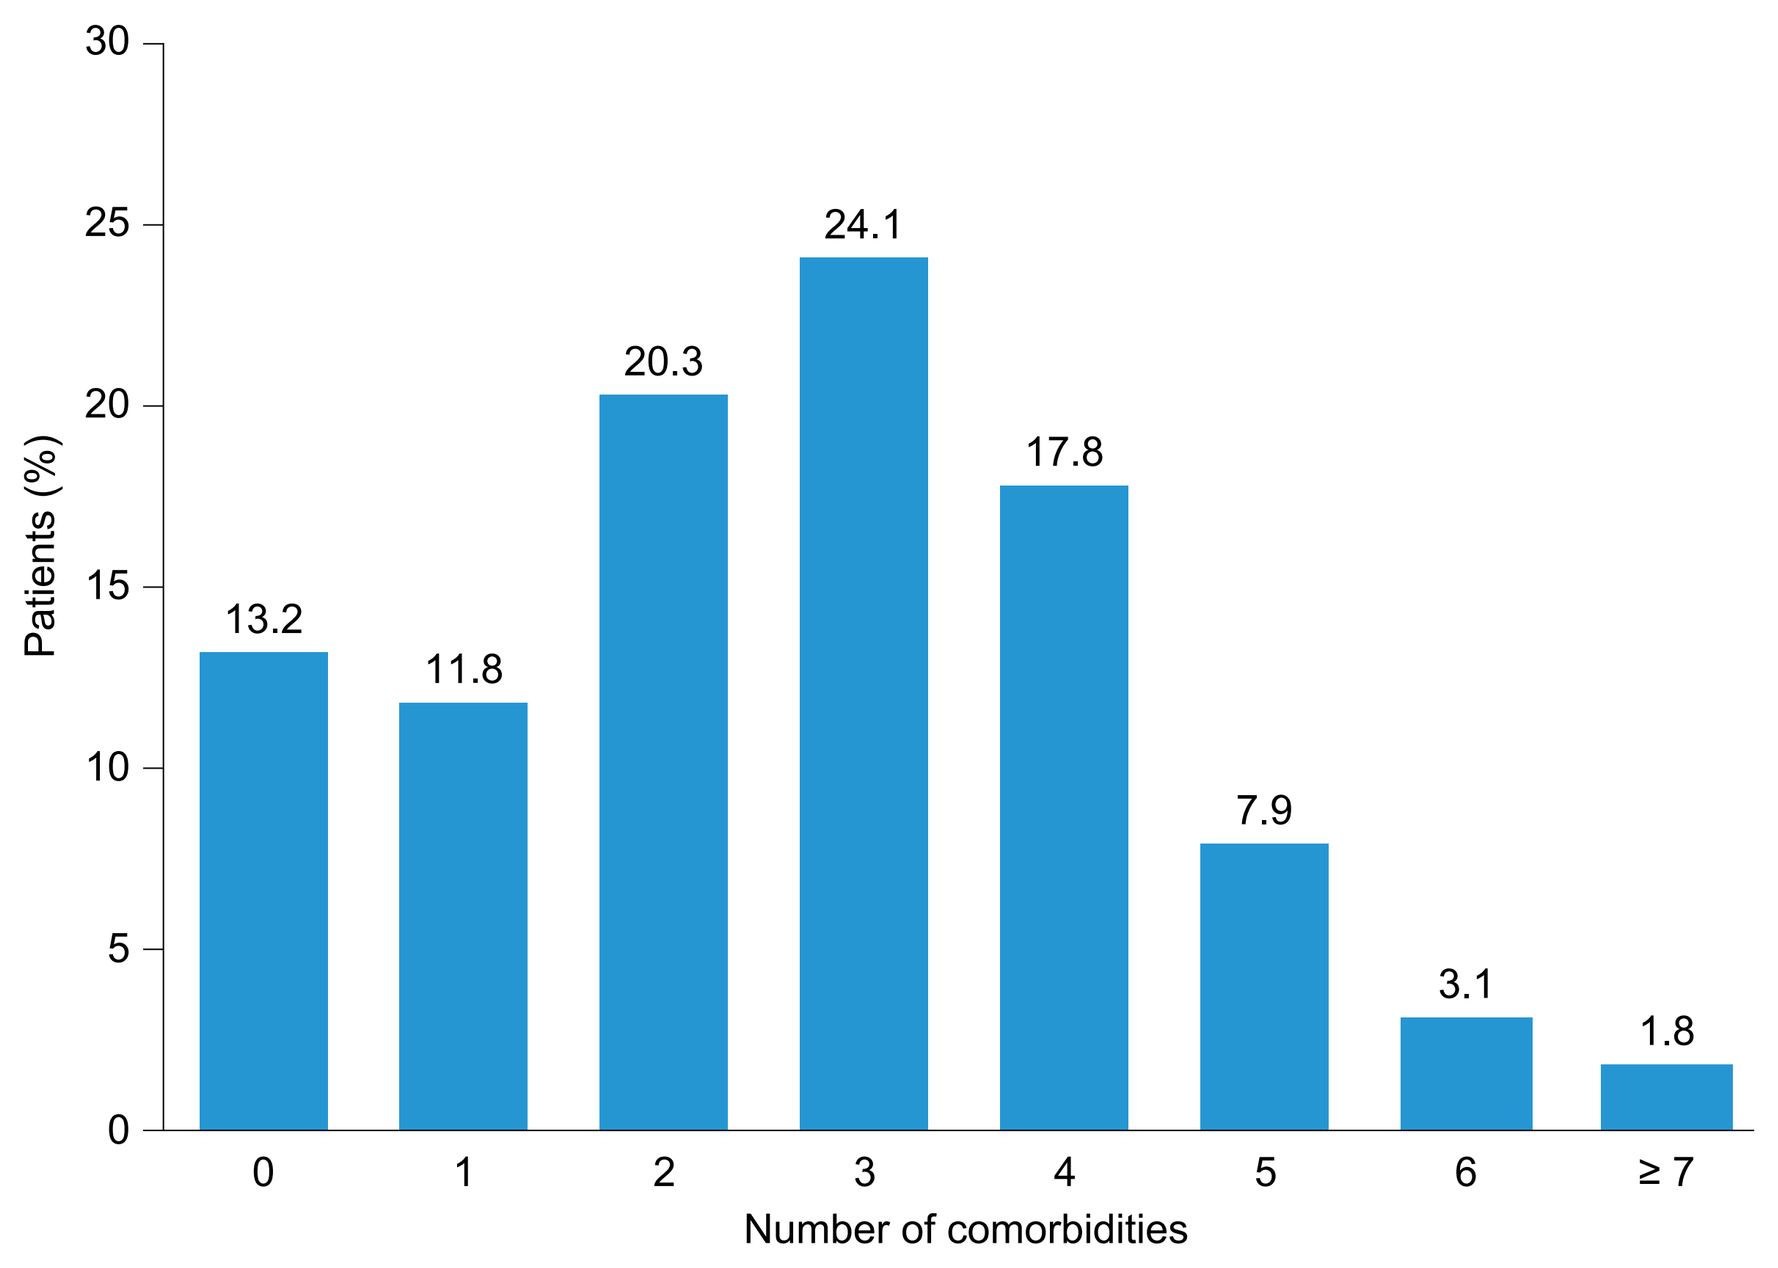
Patients (%)
30
25
20
15
10
5
0
13.2
11.8
20.3
24.1
17.8
7.9
3.1
1.8
0
1
2
3
4
5
6
≥ 7
Number of comorbidities

MASH, metabolic dysfunction-associated steatohepatitis.

Table 2. Demographics and characteristics of people living with MASH who participated in qualitative interviews

Parameter	Patients with MASH (N = 14)
Sex, n (%)	
Male	2 (14)
Female	12 (86)
Age, years, mean (range)	52 (28–74)
Fibrosis stage, n (%)	
F2	6 (43)
F3	6 (43)
F4[a]	2 (14)
Current treatments, n (%)[b]	
Resmetirom	5 (36)
Incretin-based therapies	8 (57)
Lifestyle modifications	10 (71)

[a]Patients with F4 MASH were included only in the first phase of interviews.
[b]These are not mutually exclusive.
MASH, metabolic dysfunction-associated steatohepatitis.

Figure 2. Distribution of comorbidities among patients with F2/F3 MASH identified in the claims analysis

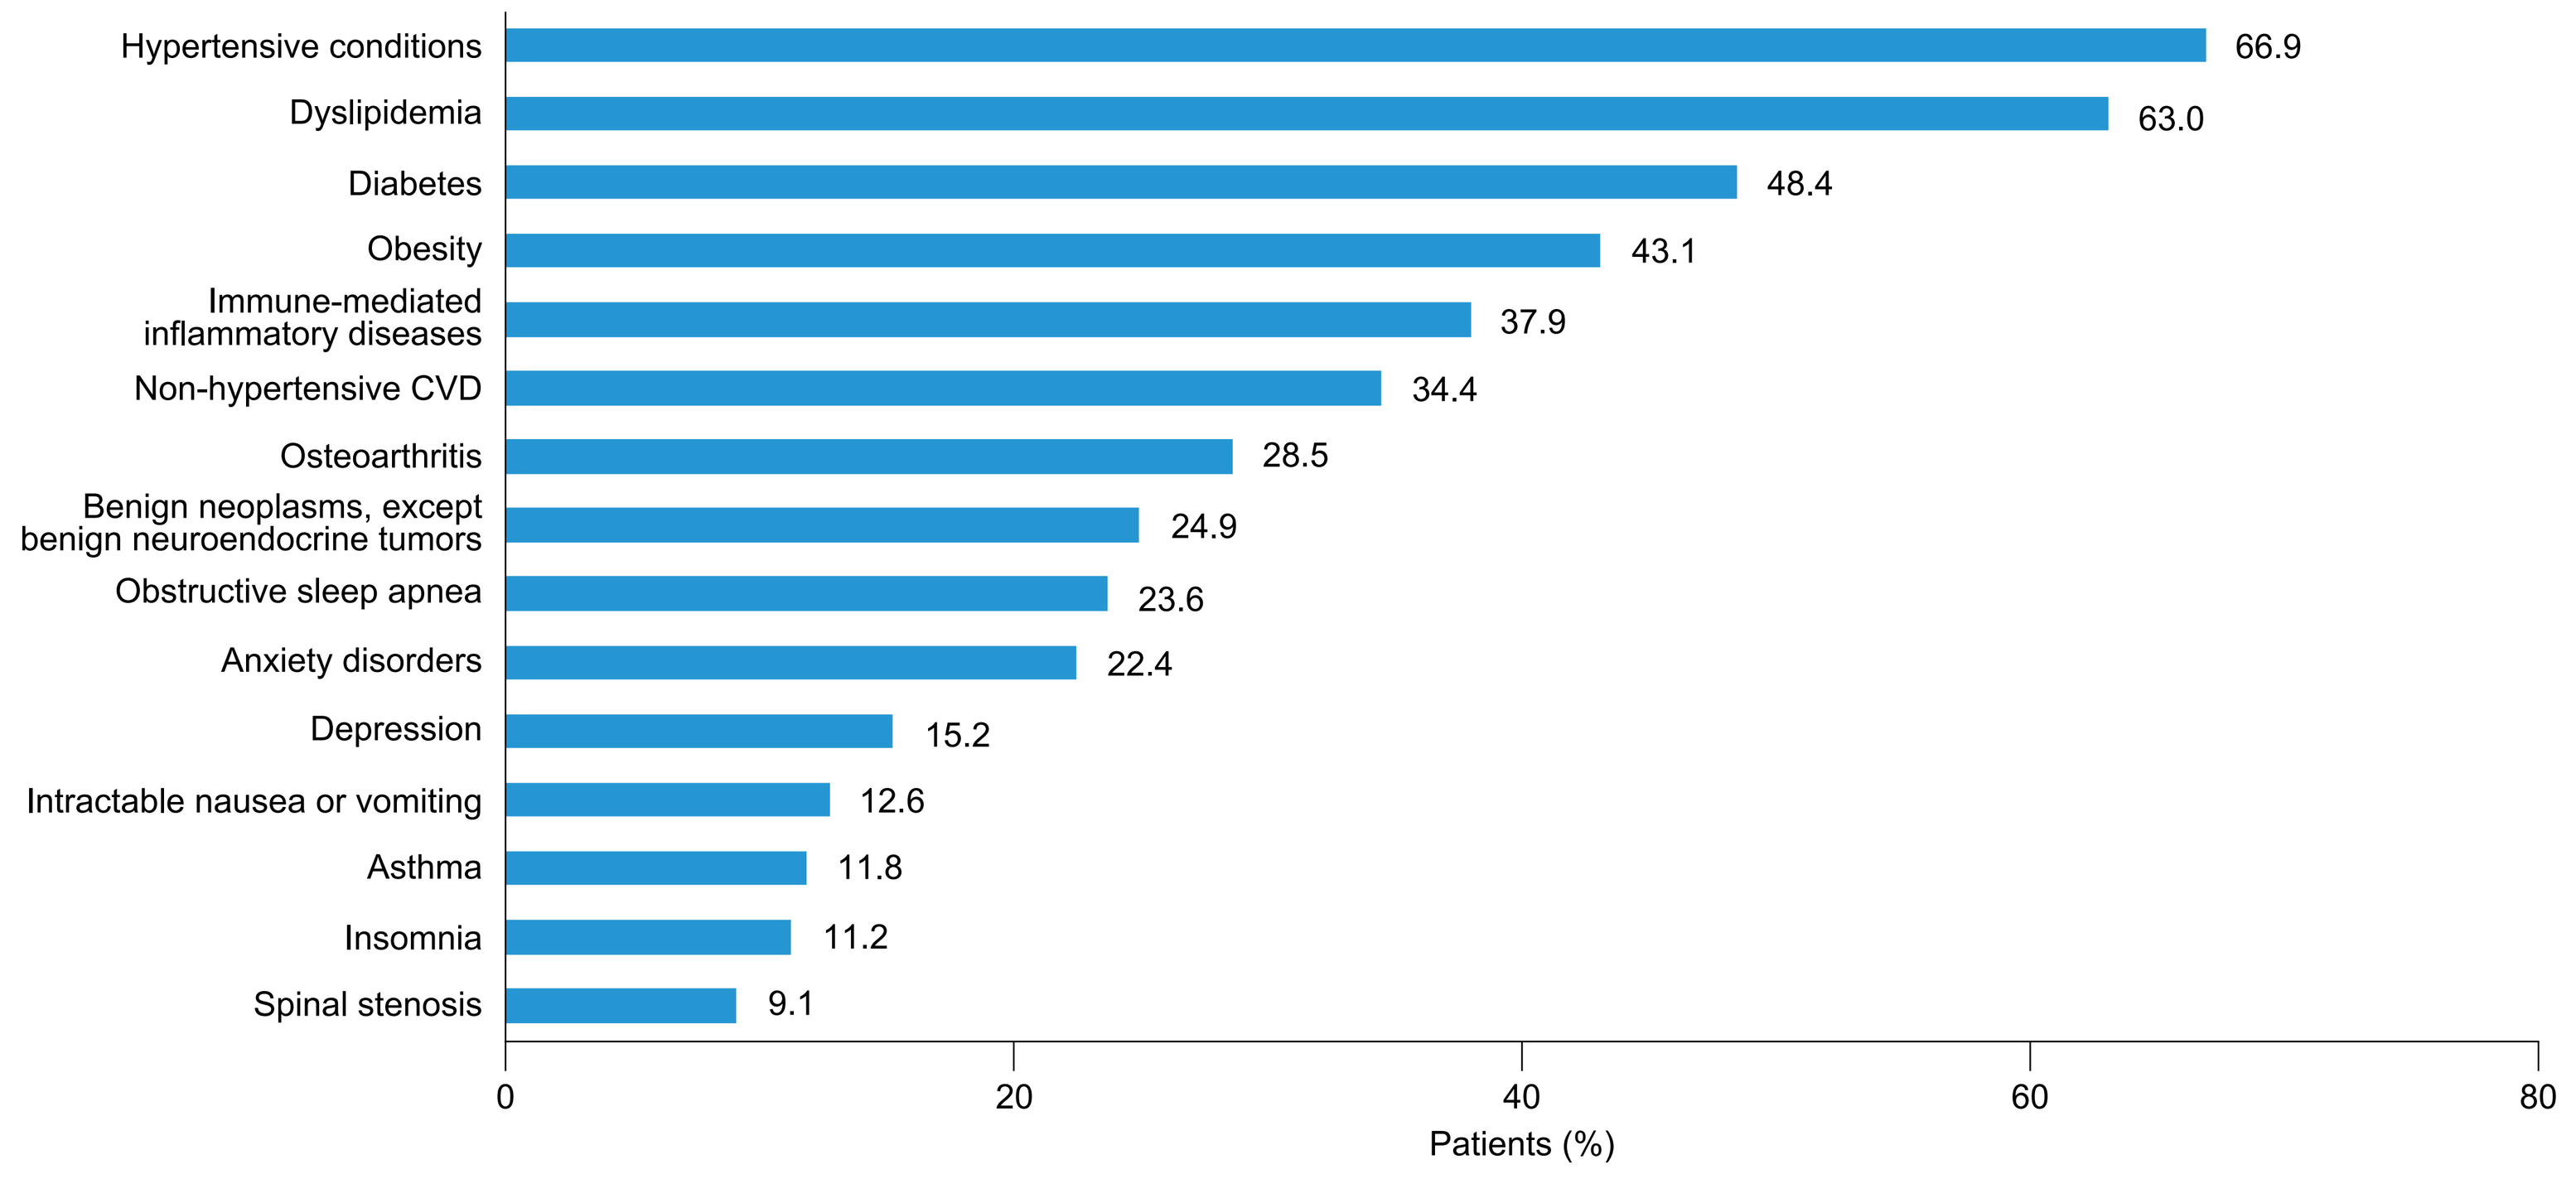
Hypertensive conditions 66.9
Dyslipidemia 63.0
Diabetes 48.4
Obesity 43.1
Immune-mediated inflammatory diseases 37.9
Non-hypertensive CVD 34.4
Osteoarthritis 28.5
Benign neoplasms, except benign neuroendocrine tumors 24.9
Obstructive sleep apnea 23.6
Anxiety disorders 22.4
Depression 15.2
Intractable nausea or vomiting 12.6
Asthma 11.8
Insomnia 11.2
Spinal stenosis 9.1
0
20
40
60
80
Patients (%)

CVD, cardiovascular disease; MASH, metabolic dysfunction-associated steatohepatitis.

MASH diagnosis is often delayed and overshadowed by comorbidities

- Many patients felt they were diagnosed 'late' or after their fibrosis had already progressed significantly.
- Some patients felt that the urgency of their MASH diagnosis was affected by 'weight bias'.

> *"I really did feel like even [in] my primary care they looked at me and said 'Oh there's just another fat person'."*

- Many patients were advised only to diet and exercise, which felt dismissive, especially for those struggling with weight loss.
- Other comorbidities were often prioritized first, with abnormal liver findings discovered incidentally.

> *"It was largely by accident. I was undergoing some tests for something completely unrelated and one of the results, I don't even remember which, but one of the results came back a little bit odd."*

Patients can find the diagnosis of MASH to be confusing

- Many patients had never heard of MASH, and some felt confused when 'fatty liver' 'suddenly' became MASH.
- Patients noted that specialists may provide incomplete or simplistic explanations of MASH and potentially downplay its seriousness or urgency.

> *"I was just listening to what he was saying, which wasn't a lot. Just very nonchalant. 'It's no big deal. I see this, that and the other all the time.' He didn't really elaborate on stage 3 fibrosis. All of this I researched when I got home."*

> *"I don't feel like she was very empathetic … I felt like she felt like it was no big deal … [I was] an inconvenience."*

- Visuals, models, printed materials, and online resources, as well as advocacy groups, helped patients understand MASH.

Patients have multiple comorbidities, which complicate focus

- Patients reported living with a variety of other conditions, including hypertension, dyslipidemia, type 2 diabetes, obesity, and cancer.

> *"I've documented 27 other medical conditions. I'm on 18 pills a day."*

- Some patients deprioritized their MASH diagnosis and management owing to competing demands from other conditions.

> *"I think what I largely did was I took the issues that were most pressing at that moment, and as I started getting treatment for one condition after another."*

> *"My focus primarily ... was dealing with cardiac issues because that was a bigger deal at the time than any gastrointestinal, liver, gall bladder issues."*

- Patients with family history of liver disease were more likely to prioritize MASH.

Access to MASH medication may be inconsistent

- Diet and exercise were the most common initial treatments for patients' MASH.
- Incretin-based therapies were often started for comorbidities before diagnosis of MASH.
- Some patients experienced barriers to accessing medications for MASH (e.g. cost, insurance issues).

Patients often carry the burden of coordinating their own care

- Many patients needed to coordinate their own care (e.g. scheduling follow-ups and confirming communication between specialists).

> *"They're aware to some extent, for sure, but I don't know how extensively they talk to one another …"*

- Patients needed to keep track of medications for different comorbidities themselves.

> *"It's so hard to juggle so many different health conditions and medications."*

> *"Right now the first thing I do anytime anyone mentions a new drug is I start looking up interactions with the stuff I'm already on."*

- Patients reported feeling overwhelmed by their routine health maintenance, with MASH monitoring adding to their burden.

> *"I had to go see a specialist who lived an hour and 45 minutes away."*

> *"I'm just frustrated because you have to see every specialist and it takes its toll. It takes its toll."*

DISCUSSION

- The high incidence and type of comorbidities reported in our study is consistent with published epidemiology studies.[6,7]
- Diagnostic delays, weight bias, minimization, and competing priorities are challenges in the management of MASH.
- Treatment options for some comorbidities may counteract treatment goals in others (e.g. antidepressants and systemic corticosteroids may counteract weight loss),[8-10] emphasizing the need for coordinated care.
- Overall, the results of our study highlight the need for holistic, integrated, informed, and empathetic care for people living with MASH.

CONCLUSIONS

- People living with MASH alongside multiple comorbidities face substantial burdens that extend beyond symptoms.
- This highlights the need to educate and support people at risk of MASH progression beyond lifestyle and comorbidity management.

FUNDING

This study was funded by 89bio, San Francisco, CA, USA.

CONFLICTS OF INTEREST

Jamie Hadac and Leslie Fok are employees and were stockholders of 89bio. Ashwin Anand, Daniel Neilson, and Wouter van der Pluijm are employees of Forian and received funding from 89bio to conduct these analyses. Julia Jones and Audrey Wu are employees of ThinkGen and received funding from 89bio to conduct these analyses.

ACKNOWLEDGMENTS

The authors would like to thank the patients who participated in the interviews. Medical writing support was provided by Sarah Graham, PhD, of Oxford PharmaGenesis, Melbourne, Australia, and funded by 89bio.

REFERENCES

1. Chalasani N *et al. Hepatology* 2018;67:328–57; 2. Younossi ZM *et al. Hepatology* 2023;77:1335–47; 3. Rinella ME *et al. Hepatology* 2023;77:1797–835; 4. Nielsen EM *et al. J Gen Intern Med* 2022;37:2587–90; 5. Forian. CHRONOS™. 2025. Available from: https://forian.com/chronos/ (Accessed October 23, 2025); 6. Glass LM *et al. Fed Pract* 2019;26:64–71; 7. Tapper EB *et al. Gastro Hep Advances* 2022;1:1049–87; 8. Durell N *et al. J Pharm Technol* 2022;38:283–8; 9. Mouawad M *et al. Arch Clin Biomed Res* 2025;9:183–95; 10. Kulkarni S *et al. BMJ Open* 2022;12:e061476.

Presented at the 23rd World Congress Insulin Resistance Diabetes & Cardiovascular Disease (WCIRDC) 2025, December 3–6, 2025, Los Angeles, CA, USA